Historical and Contemporary Burden of End-Stage Kidney Disease Among Torres Strait Islander People of Australia, 1963-2022

Prof Jaqui Hughes[1], Ms Katie Cundale[1], Assoc Prof Shahid Ullah[2], Assoc Prof Karla J Canuto[1]

[1] College of Medicine and Public Health, Flinders University, Darwin, NT
[2] College of Medicine and Public Health, Flinders University, Adelaide, SA

Background: What we know

Torres Strait Islander people of Maluilgau – the near western island cluster of the Torres Strait – have lived and thrived on their lands and waters for thousands of years.

Diabetes is a new disease in this time-continuum, and because it causes progressive disease (complications) in many body parts, it also causes disability and reduced survival, which in the Torres Strait, impacts people at the individual, family, and community level.

Hospital admission for diabetes-related amputation has been known in the Maluilgau region since 1962. Kidney-failure is another late-stage diabetes complication, and community-accessible haemodialysis infrastructure in Badu Island was proposed by community to be locally accessible support for Maluilgal (the peoples of Maluilgau). Kuiku Mabaigal (head people) of Maluilgau have gained Federal Government support to scope out the design of a 4-chair satellite haemodialysis facility.

Aim: What we need to find out

Kuiku Mabaigal need information from Australia and New Zealand Dialysis and Transplant (ANZDATA) Registry about the burden of kidney failure among Torres Strait Islander peoples, to assist Elders' role in safe-guarding community-wellbeing and survival.

Methods: What we know

We requested de-identified health information from the ANZDATA Registry to describe when, where, who, why, and how Torres Strait Islander people had received dialysis and transplant care in Australia during 1966-2022.

The information request included de-identified demographic details (age, sex, location), and information about primary disease, comorbidities, and treatment modality data for adults (≥18 years old) who commenced kidney replacement therapy (KRT) in Australia between 1966 and 2022. Torres Strait Islander peoples in ANZDATA were identified if an ANZDATA ethnicity identifier code was recorded as "Torres Strait Islander". Patient survival outcomes were assessed in Cox proportional hazards models and survivals were depicted in Kaplan–Meier survival curves.

This research activity and its conduct was approved by Maluilgal Tribal Governance and an Indigenous Health Ethics Committee (Aboriginal Health Research Ethics Committee, of the Aboriginal Health Council of South Australia. AHREC Protocol #: 04-23-1086).

ANZDATA is physically located on Kaurna Yarta (Adelaide, Australia), and is a Clinical Quality Registry, working with renal units in Australia and New Zealand. It maintains a record of patients who have kidney failure and receive dialysis and transplant care from an Australian renal unit. ANZDATA routinely collates health outcomes data, to report on the amount, type and quality of KRT routinely provided in Australian health care facilities. ANZDATA has been the custodian of collated information since 1966. Torres Strait Islander people worked with ANZDATA for a number of years, previously contributing to the ANZDATA "Guidance on data requests relating to patient ethnicity" policy, published in 2020. See www.anzdata.org.au/anzdata/services/data-policies for more information.

Results: What we found out

Torres Strait Islander people have commenced kidney replacement therapy at an increasing rate over time, with most people utilising haemodialysis.

Figure 1. Torres Strait Islander adults starting dialysis during 1963-2022 (ANZDATA, 2023)

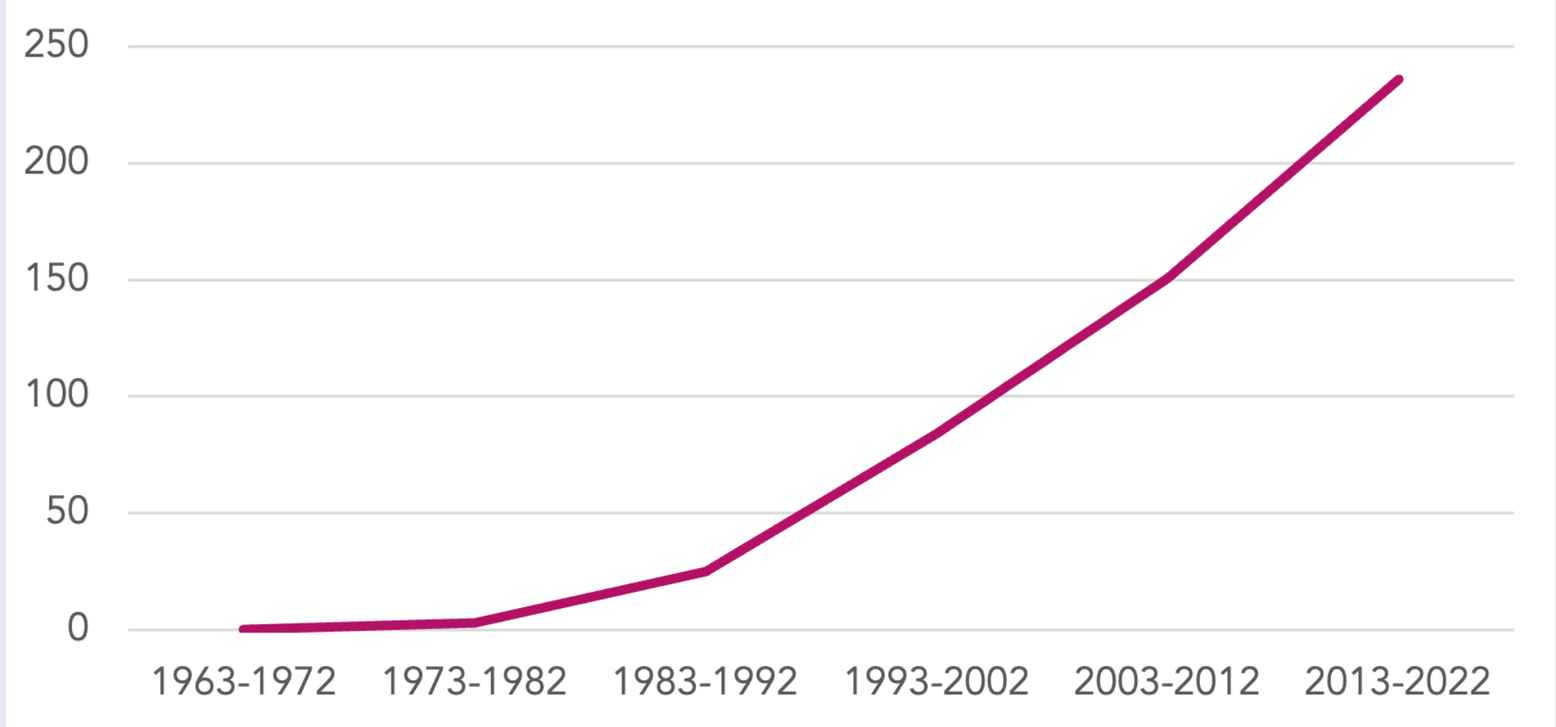
250
200
150
100
50
0
1963-1972
1973-1982
1983-1992
1993-2002
2003-2012
2013-2022

When: 1973 was the first-year dialysis treatment was provided to a person who identified as a Torres Strait Islander. ANZDATA has recorded a total of 499 Torres Strait Islander adults receiving KRT since 1973. The most recent decade (January 2013- December 2022, compared to earlier decades) recorded the highest proportion of Torres Strait Islander adults commencing KRT (47%, (236/499)) (Figure 1).

Where: KRT care was accessed by Torres Strait Islander adults across Australia, most commonly in outer regional (49%, 235/499) and very remote locations (35%, 165/499). The State of Queensland provided care to 463 Torres Strait Islander adults — this was 93% of all Torres Strait Islander people since 1973 (Figure 2).

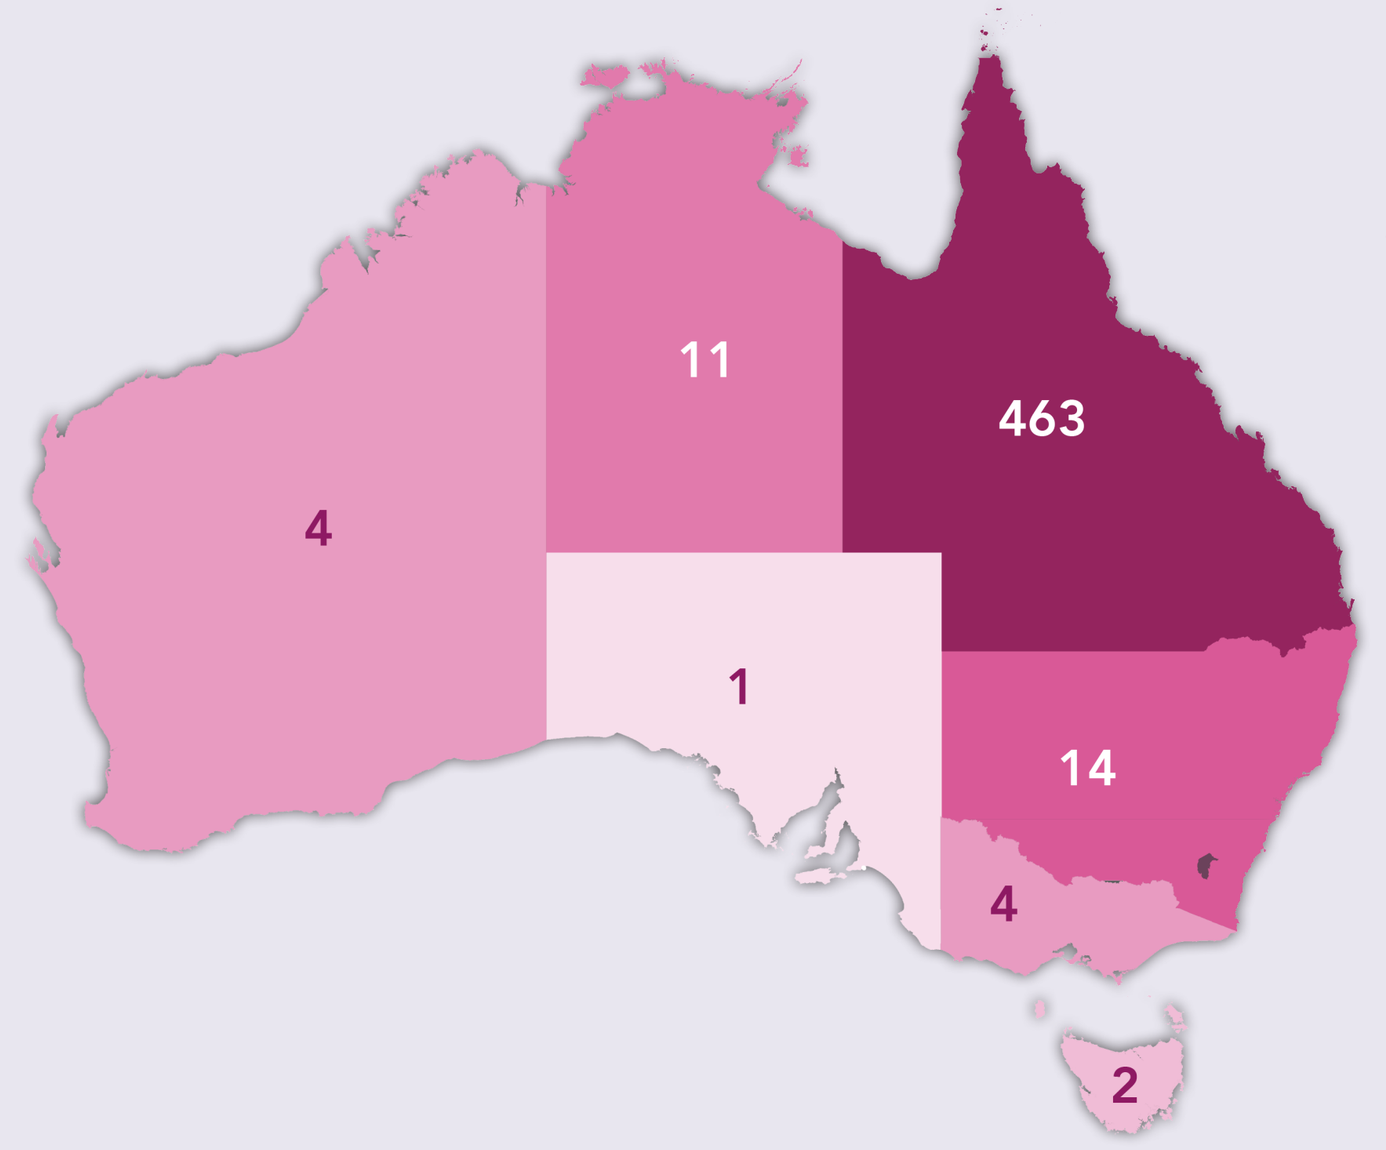
11
463
4
1
14
4
2

Figure 2. State or Territory of initial dialysis unit for Torres Strait Islander people starting dialysis, 1973-2022 (ANZDATA, 2023)

Who: Torres Strait Islander women (55%) and men (45%) received KRT; the mean age of starting KRT was 54 (SD=12, n=499) years.

Why: Diabetes was the cause of kidney failure recorded by ANZDATA in most Torres Strait Islander adults (76%, 377/499).

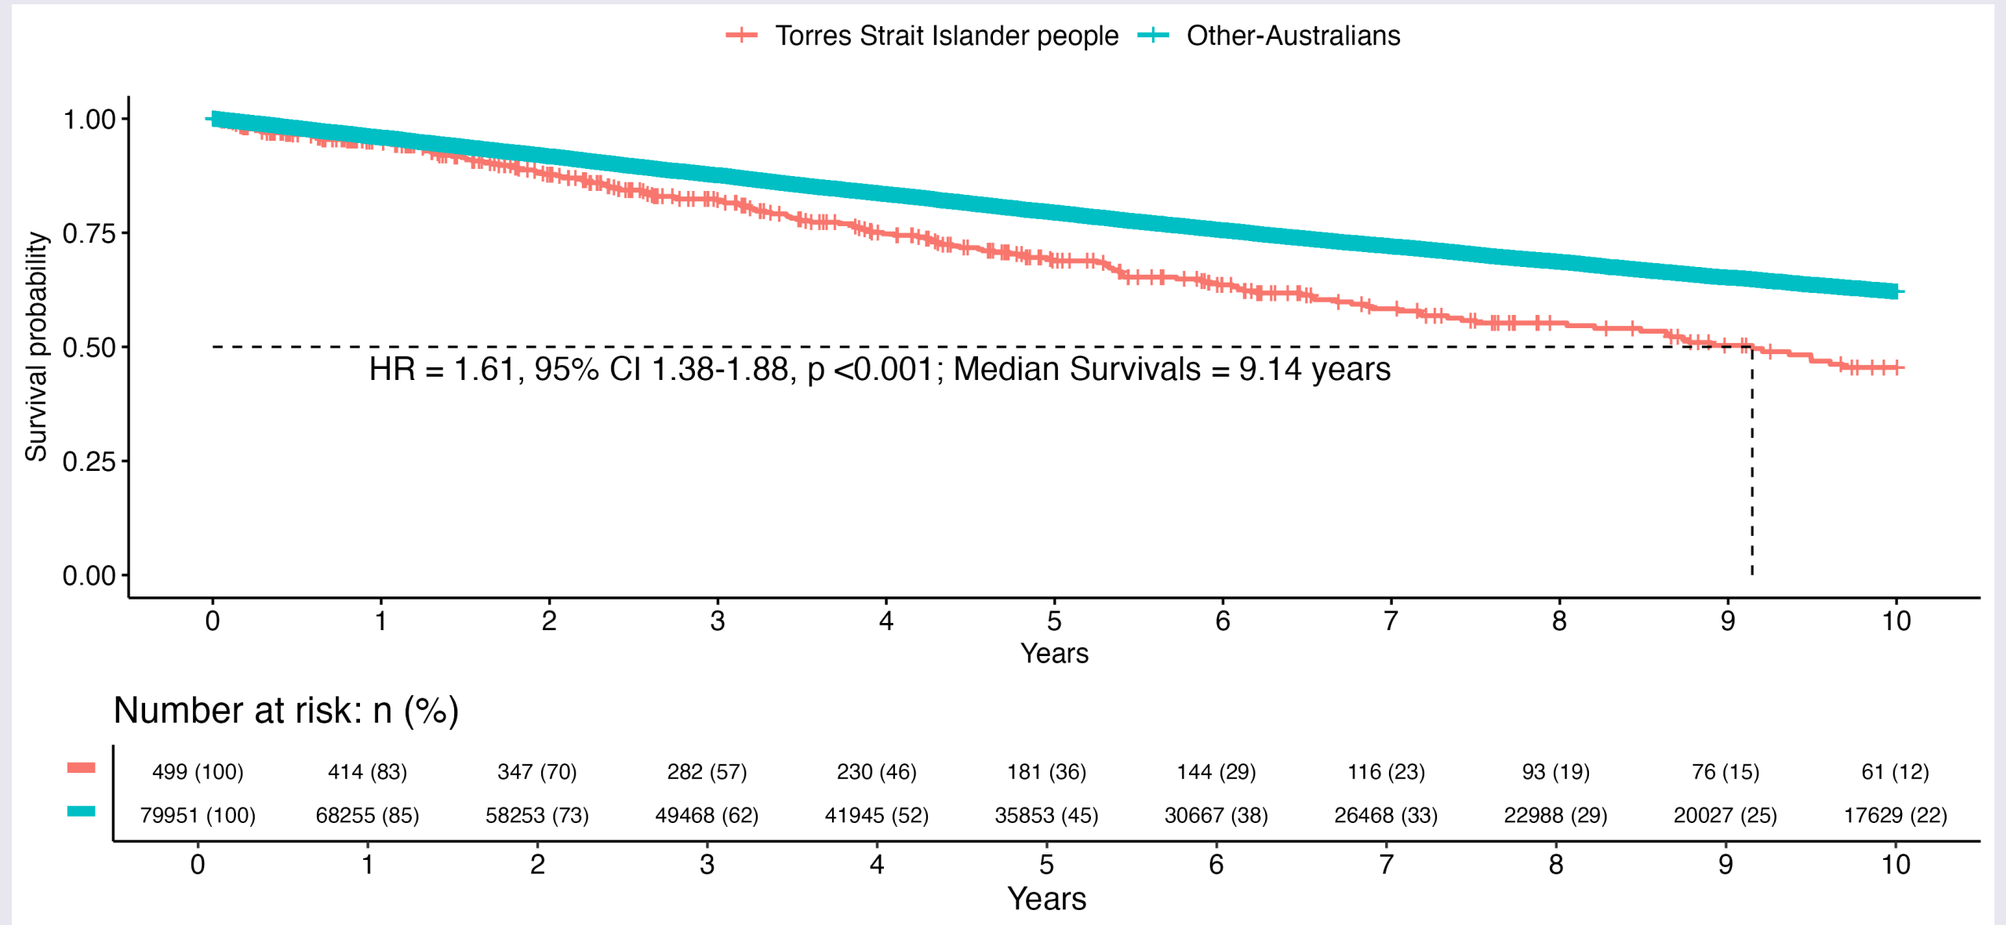
Torres Strait Islander people
Other-Australians
Survival probability
HR = 1.61, 95% CI 1.38-1.88, p <0.001; Median Survivals = 9.14 years
Years
Number at risk: n (%)
499 (100) 414 (83) 347 (70) 282 (57) 230 (46) 181 (36) 144 (29) 116 (23) 93 (19) 76 (15) 61 (12)
79951 (100) 68255 (85) 58253 (73) 49468 (62) 41945 (52) 35853 (45) 30667 (38) 26468 (33) 22988 (29) 20027 (25) 17629 (22)
Years

Figure 3. Survival probability for Torres Strait Islander people (ANZDATA, 2023)

How: Haemodialysis was the most common treatment (rather than peritoneal dialysis or transplant) over the whole period since 1973. 415 (83%) Torres Strait Islander people used haemodialysis at their first treatment. At the end of 2022, Torres Strait Islander people accessed their haemodialysis at a satellite facility (51%), or a hospital facility (26%). The first kidney transplant recorded for a Torres Strait Islander adult was in 1978. There were forty-nine transplants (49/499, 10%) in total recorded in ANZDATA up until December 2022. There have been no pre-emptive kidney transplants for Torres Strait Islander people recorded in ANZDATA.

What: At 10 years (Figure 3), survival rates for Torres Strait Islander people were lower at 50% compared to 63% for other Australians (HR = 1.61; 95% CI = 1.38-1.88, p<0.001). The median survival for Torres Strait Islander people was around 9 years. The leading causes of death was cardiovascular disease (43%, 135/499) followed by infections (21%, 65/499).

Discussion: What this means

We confirm 499 Torres Strait Islander adults had kidney failure and were supported with kidney replacement therapy across Australia, since 1973.

Kidney failure occurred as a late complication of diabetes in 76% of people, affected men and women in middle-aged years, who survived for 9 years, with satellite dialysis models of care.

Knowing how care has been provided and accessed by Torres Strait Islander people now reveals a wide opportunity for community-championed programs that will benefit the whole community — through diabetes prevention, and programs supporting wellbeing for people living with kidney disease.

Torres Strait Islander Elders sought a research partnership with clinician-academics and scientists to drive this enquiry, which establishes a baseline monitoring and evaluation health statistic.

These data may be underestimating the impact of kidney failure in Torres Strait Islander people, as ANZDATA records health outcomes of patients once accepted (by the individual and the health team) to dialysis care.

Find out more

Learn more about the Kikirriu Dan Walmai Project, which is working to bring dialysis care to Badu Island in the Torres Strait, by scanning the QR code.

Kikirriu Dan Walmai means *Our life is resurrected through overcoming sickness* in the Kala Lagaw Ya language spoken in western and central island regions of the Torres Strait.

KDW project team acknowledge the traditional owners of the Kaurareg, Larrakia and Kaurna lands on which we live and work, and the traditional owners of Maluilgau, who have championed this research activity.

We also acknowledge the multidisciplinary and medical clinicians who provide high quality culturally safe kidney clinical care to Torres Strait Islander families since 1973, and ANZDATA for holding this longitudinal dataset with Indigenous Data Governance, so we can understand quality of delivered health care available to our sovereign and isolated islands of Maluilgau of Kaiwalagal- Zenadh (known as Torres Strait).

Flinders University